Trends in Discharge Destinations Post Stroke Hospitalization in New Jersey

Markos Ezra, PhD, Abate Mammo, PhD , Debra Virgilio, RN, MPH, Emmanuel Noggoh, Yong Sung Lee, PhD

Health Care Quality Assessment, New Jersey Department of Health

New Jersey State Stroke Conference, *April 2, 2015*

Background

- This study is part of the ongoing health care quality assessment activities that the New Jersey Department of Health performs to provide information related to stroke patients in New Jersey.
- In New Jersey, stroke is the third leading cause of death and the major cause of chronic disability. Many post-stroke patients suffer functional disabilities and require rehabilitation services. Stroke patients can receive rehabilitation services at home or in an outpatient facility, rehabilitation hospital, or a skilled nursing facility (SNF).
- Since the New Jersey "Stroke Center Act" (P.L. 1971, c.136) on September 1, 2004, designating hospitals as primary or comprehensive stroke centers, the number of designated stroke centers has grown to 66 (out of 71). This paper studies trends in major discharge destinations of surviving stroke patients by socio-demographic characteristics.

Objectives

- The main objective of the study is to examine if designation of stroke centers has influenced discharge destinations of surviving patients.
- The study also examines the length of stay and cost of treatment for hospitalized patients.

Data and Methods

- This analysis uses the New Jersey Hospital Discharge Data Collection System (NJDDCS), and the New Jersey Acute Stroke Registry (NJASR).
- Socio-demographic characteristics of ischemic stroke (gender, age, race/ethnicity), health insurance coverage, average length of stay, and cost of treatment have been examined by discharge destination (Home, Rehab Facility). In the analysis, the category 'Discharged Home' includes those discharged to go home for self care or under care of organized home health service provider, while 'Rehabilitation' includes those discharged to rehabilitation facilities like SNF, ICF, long term care hospitals, etc.)
- Examination of the relationship between disability status (Modified Rankin Scale reported in the NJASR) and discharge destination by using a Chi-Square test have also been made using ischemic stroke patients.

Trends in 65 and Older Ischemic Stroke Patients Discharged to Rehabilitation by Race

Male

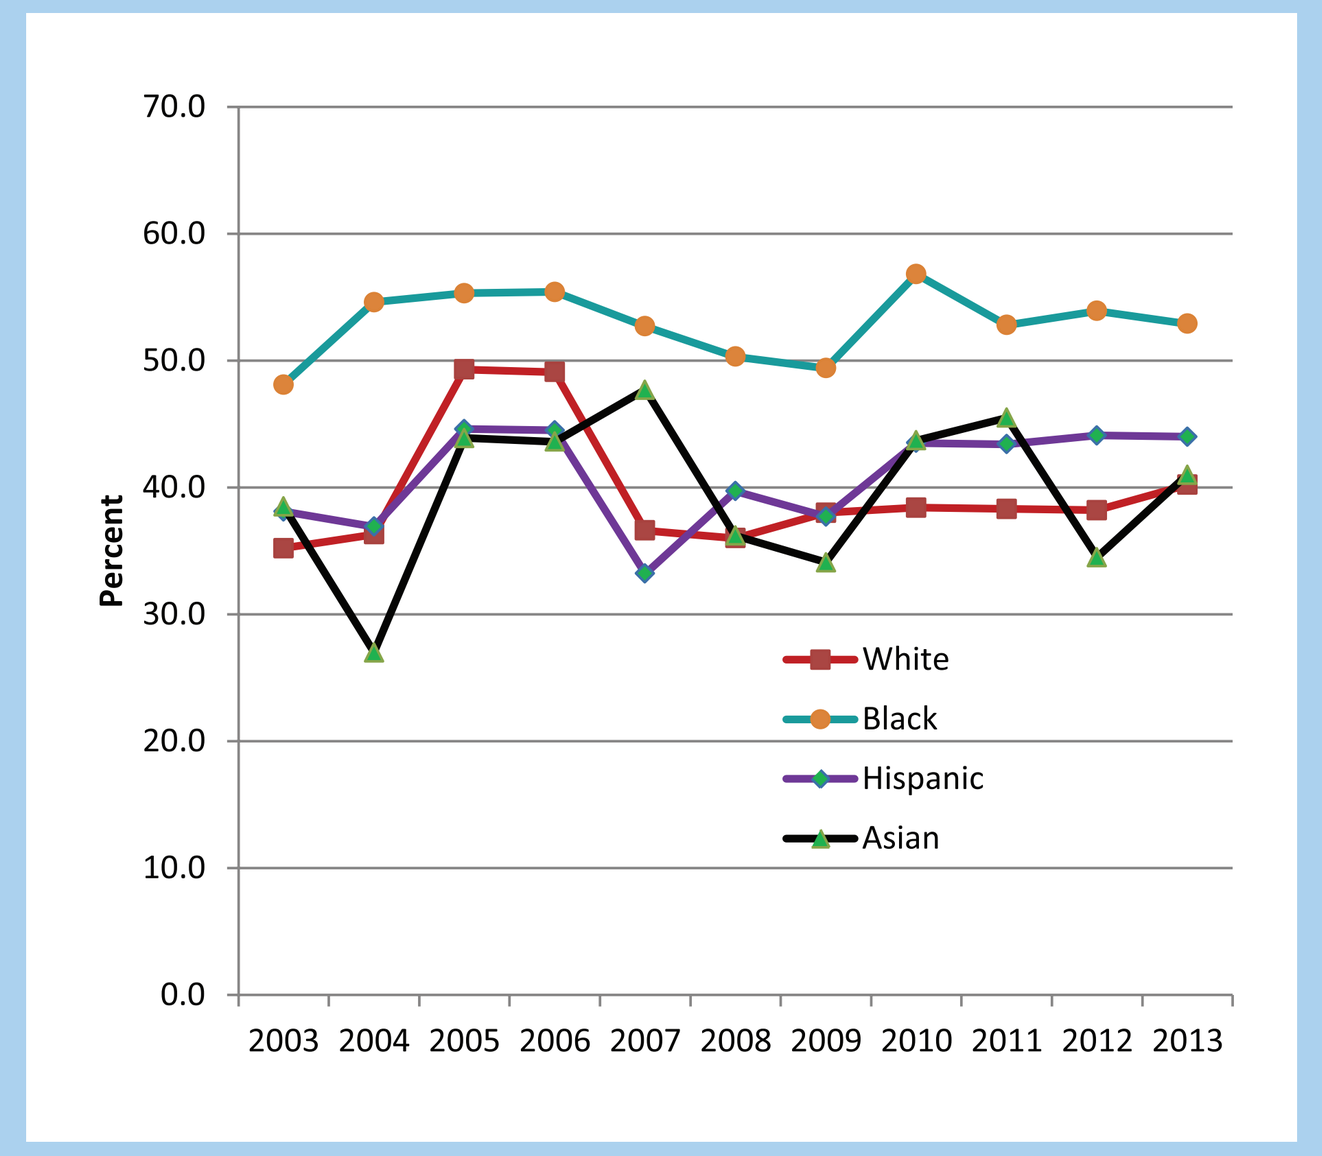

Percent
2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013
White
Black
Hispanic
Asian

Female

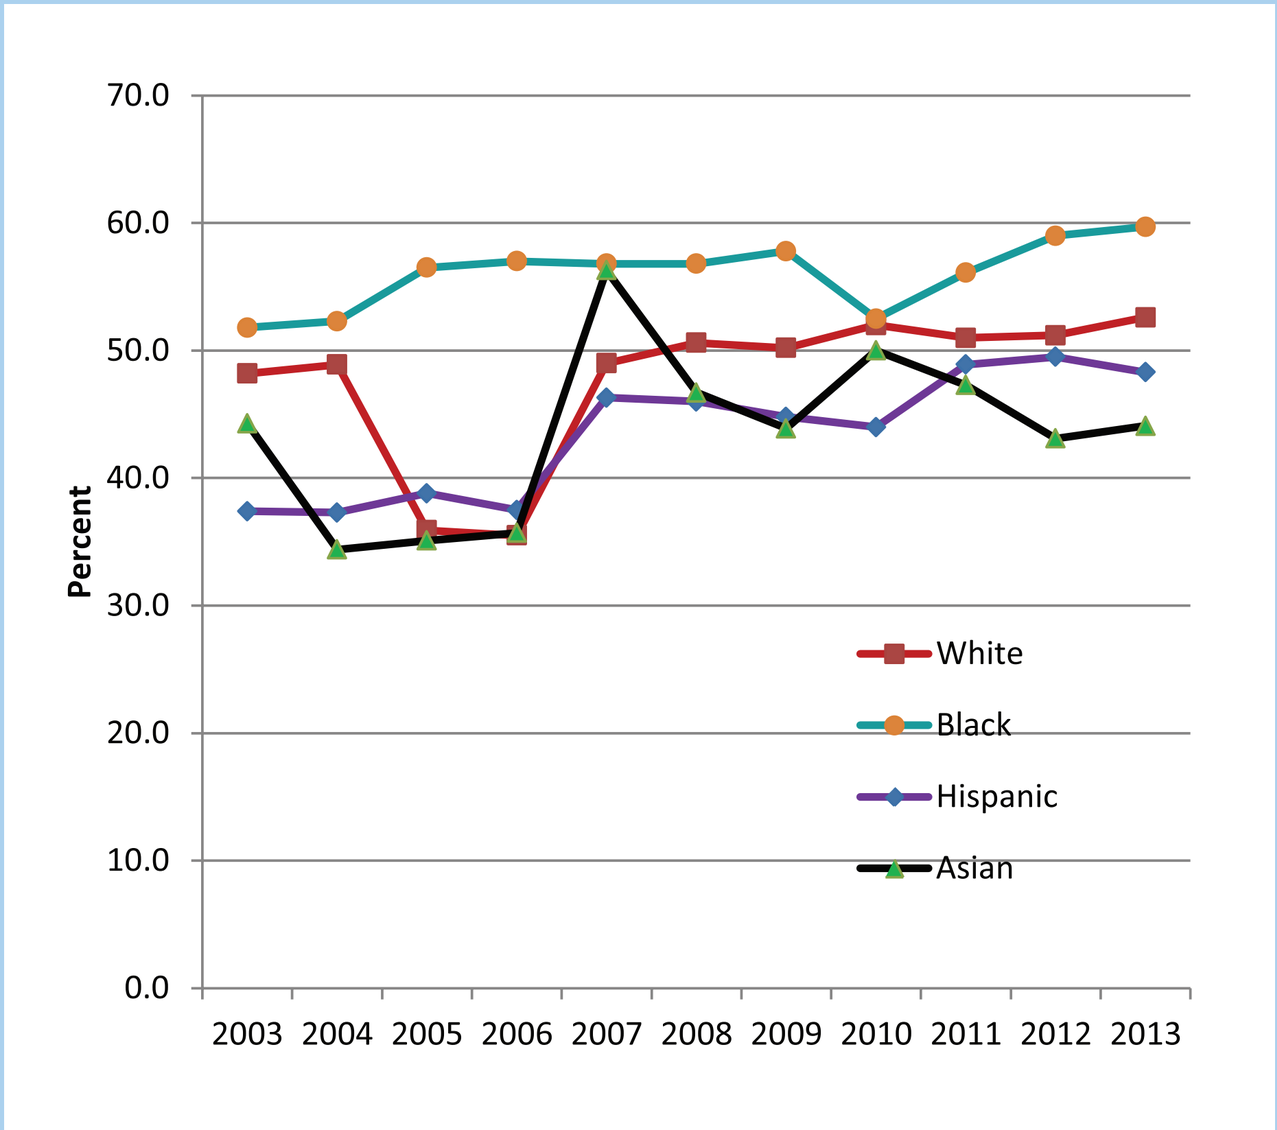

Percent
2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013
White
Black
Hispanic
Asian

Trends in 45-64 Years Old Ischemic Stroke Patients Discharged to Rehabilitation by Race

Male

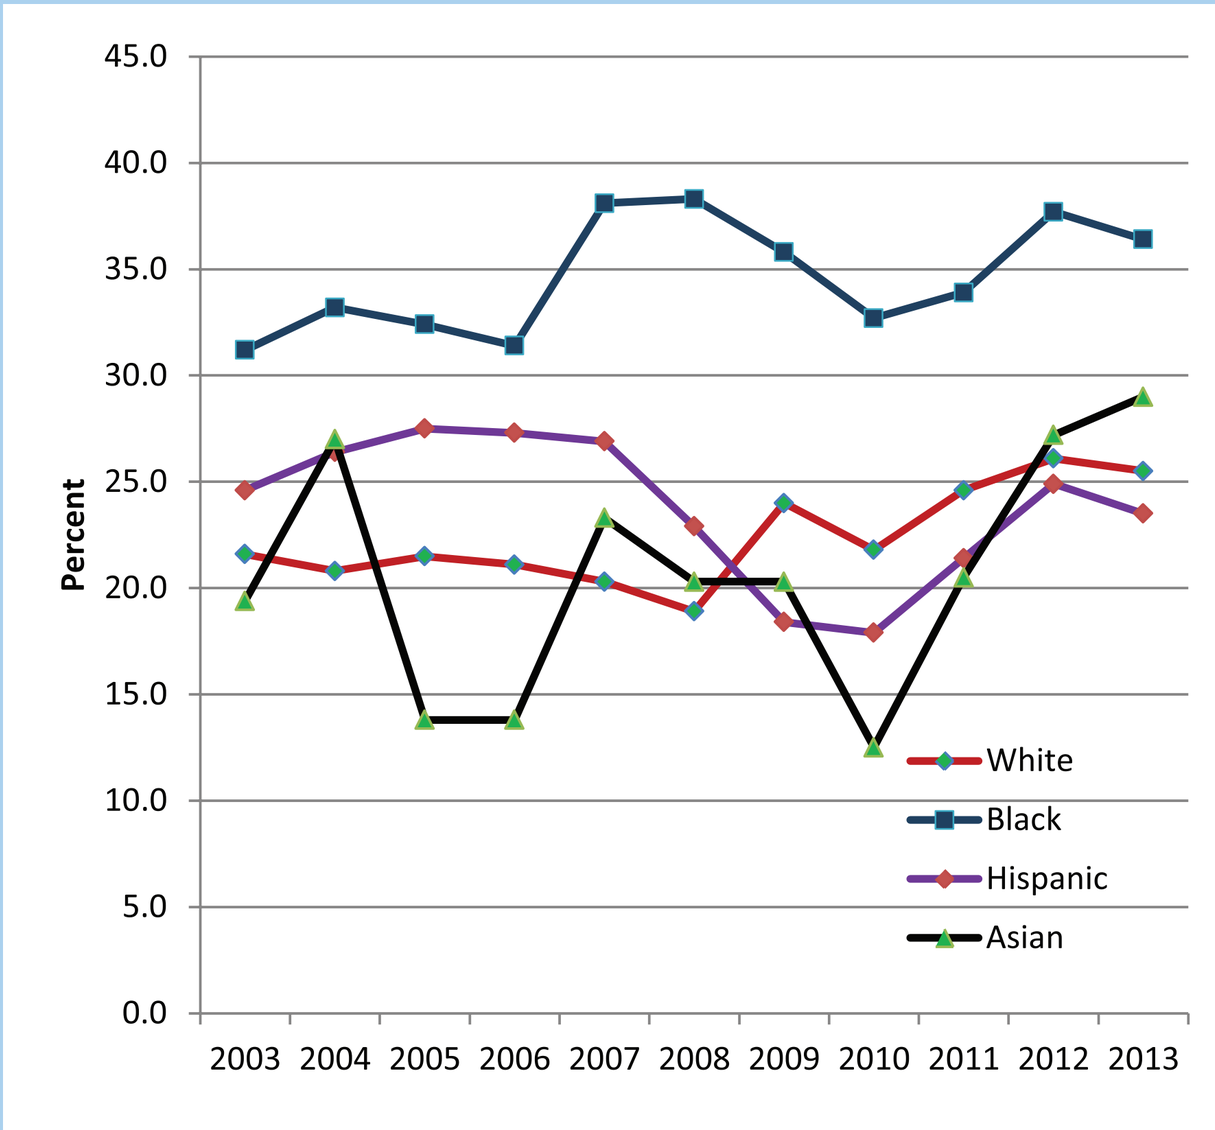

Percent
2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013
White
Black
Hispanic
Asian

Female

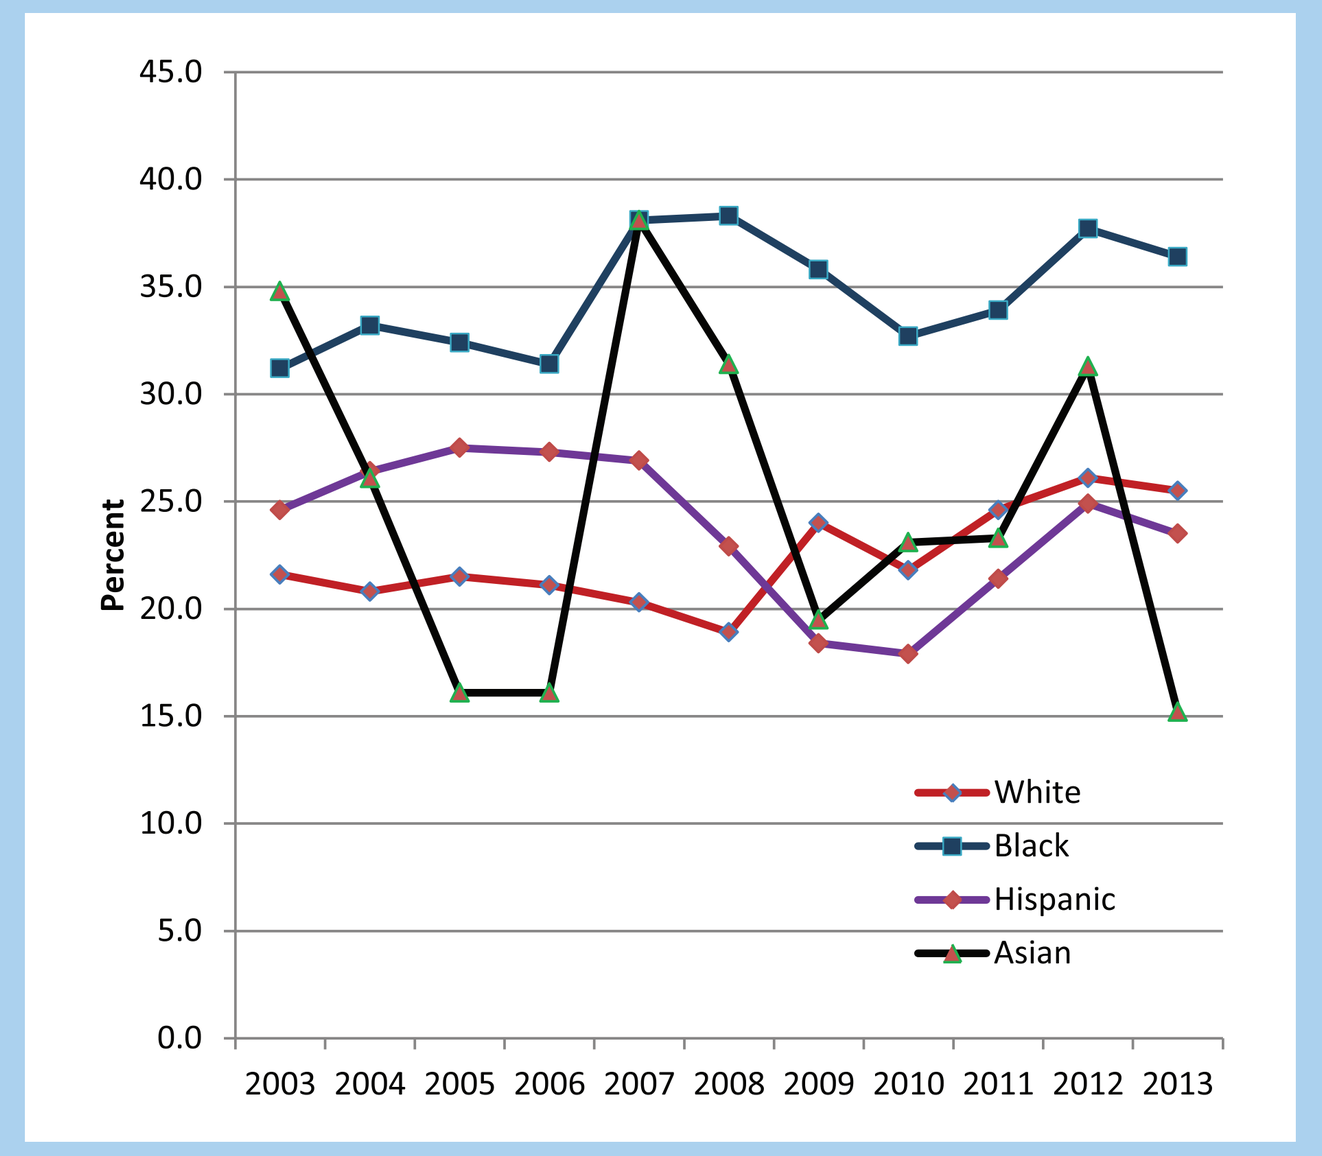

Percent
2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013
White
Black
Hispanic
Asian

Discharge Destinations of Ischemic Stroke Patients by Health Insurance Coverage: 2003 and 2013

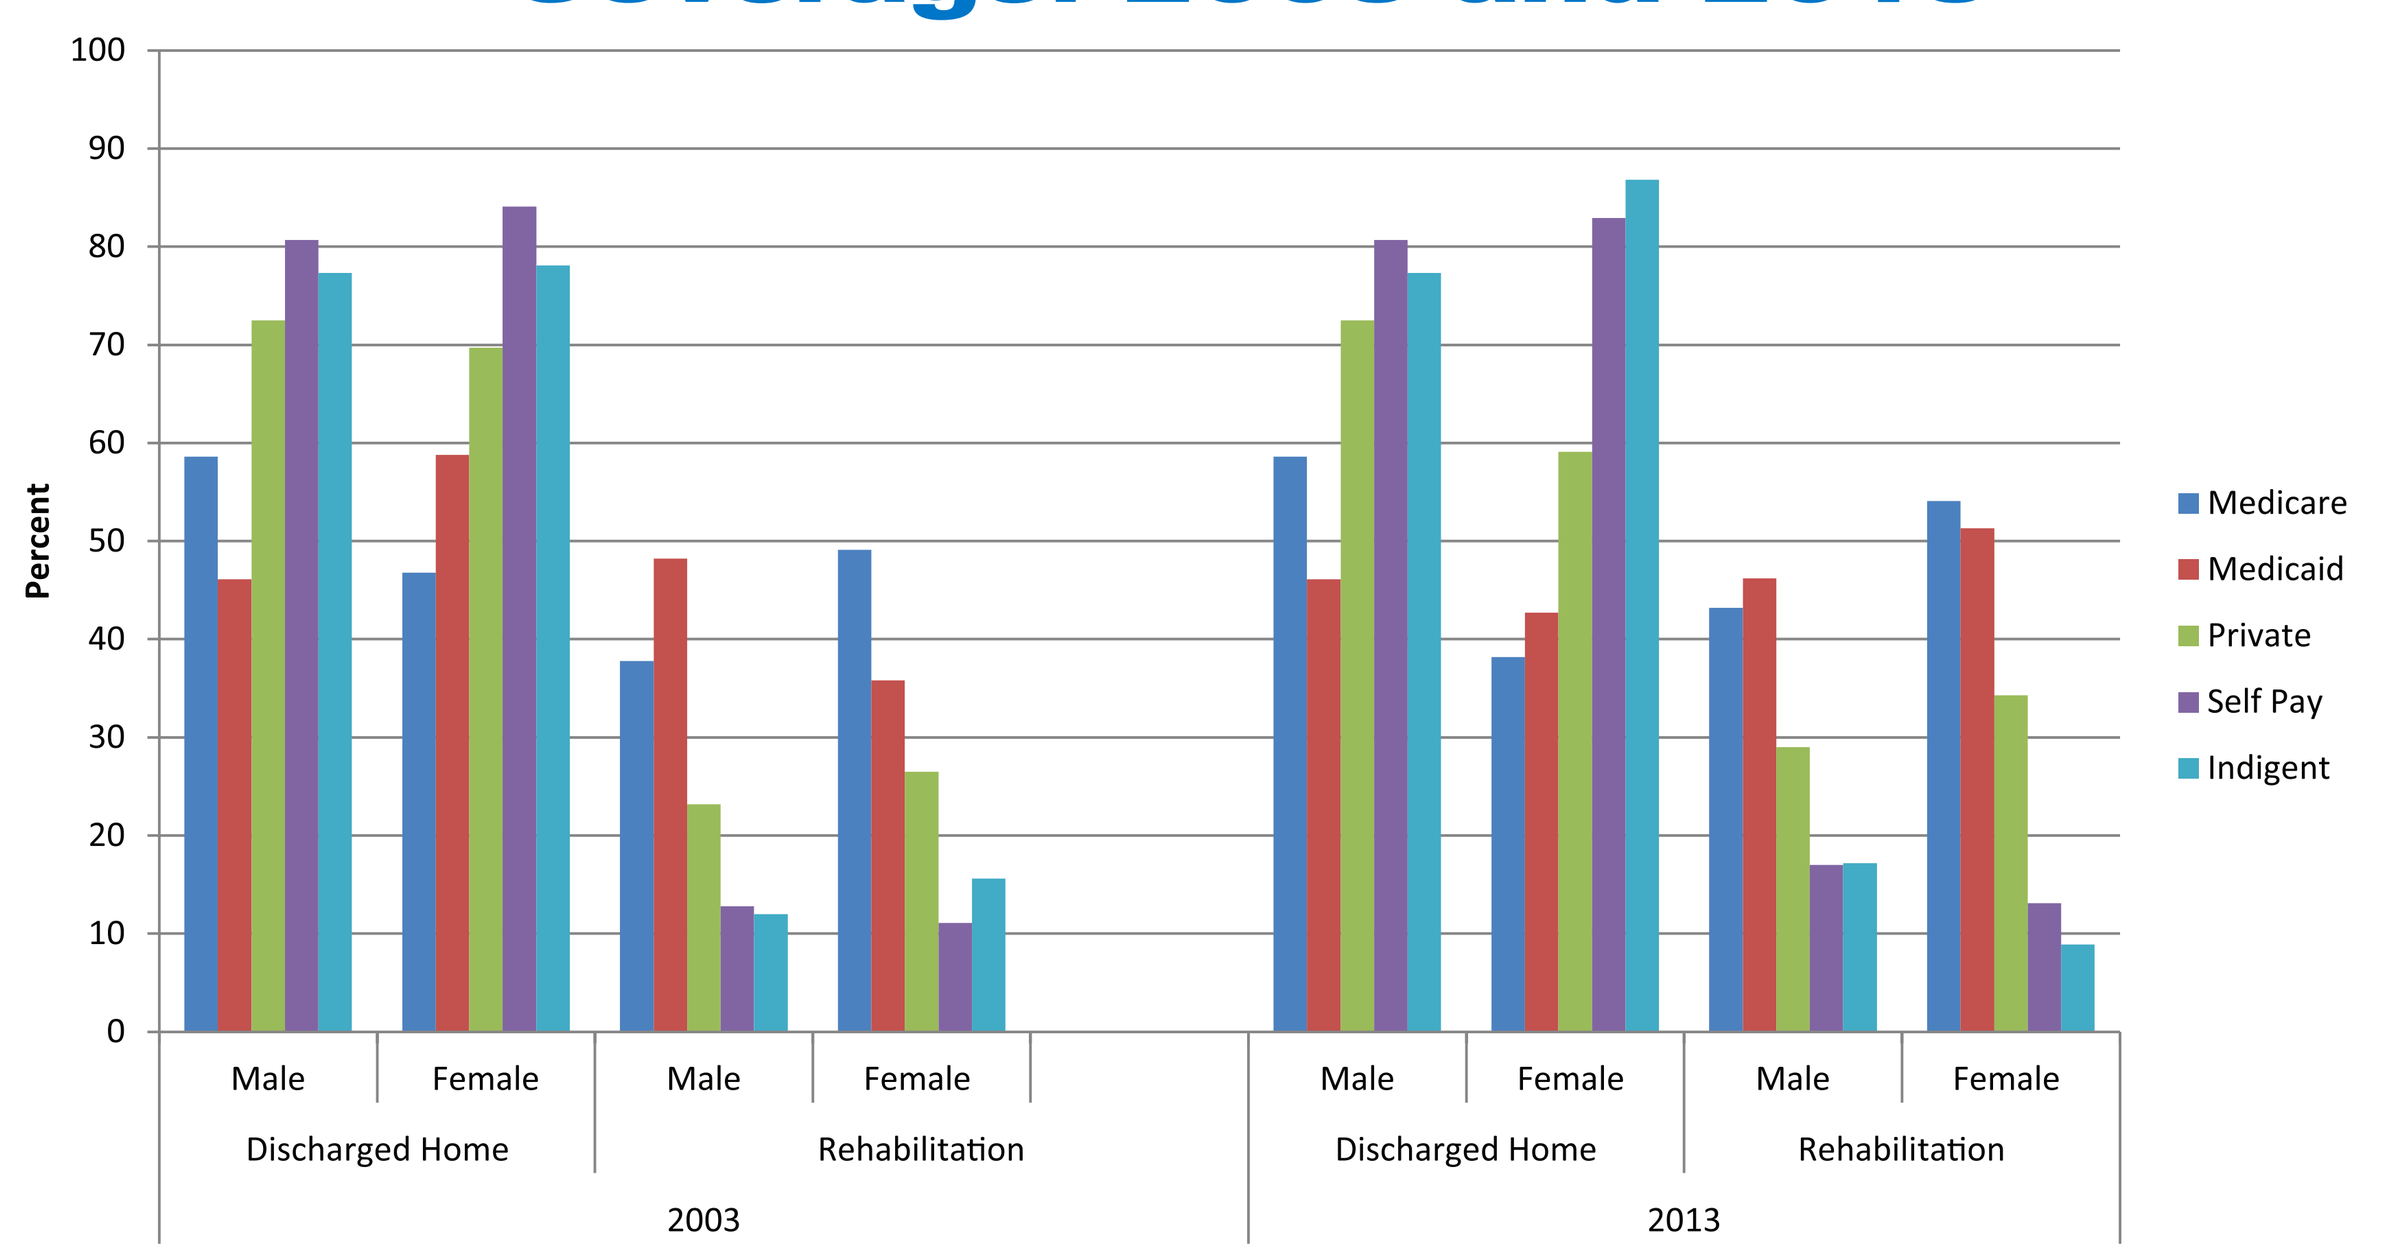

Percent
Male
Female
Discharged Home
Rehabilitation
2003
2013
Medicare
Medicaid
Private
Self Pay
Indigent

Discharge Destinations of Ischemic Stroke Patients by Race/ethnicity: 2003 and 2013

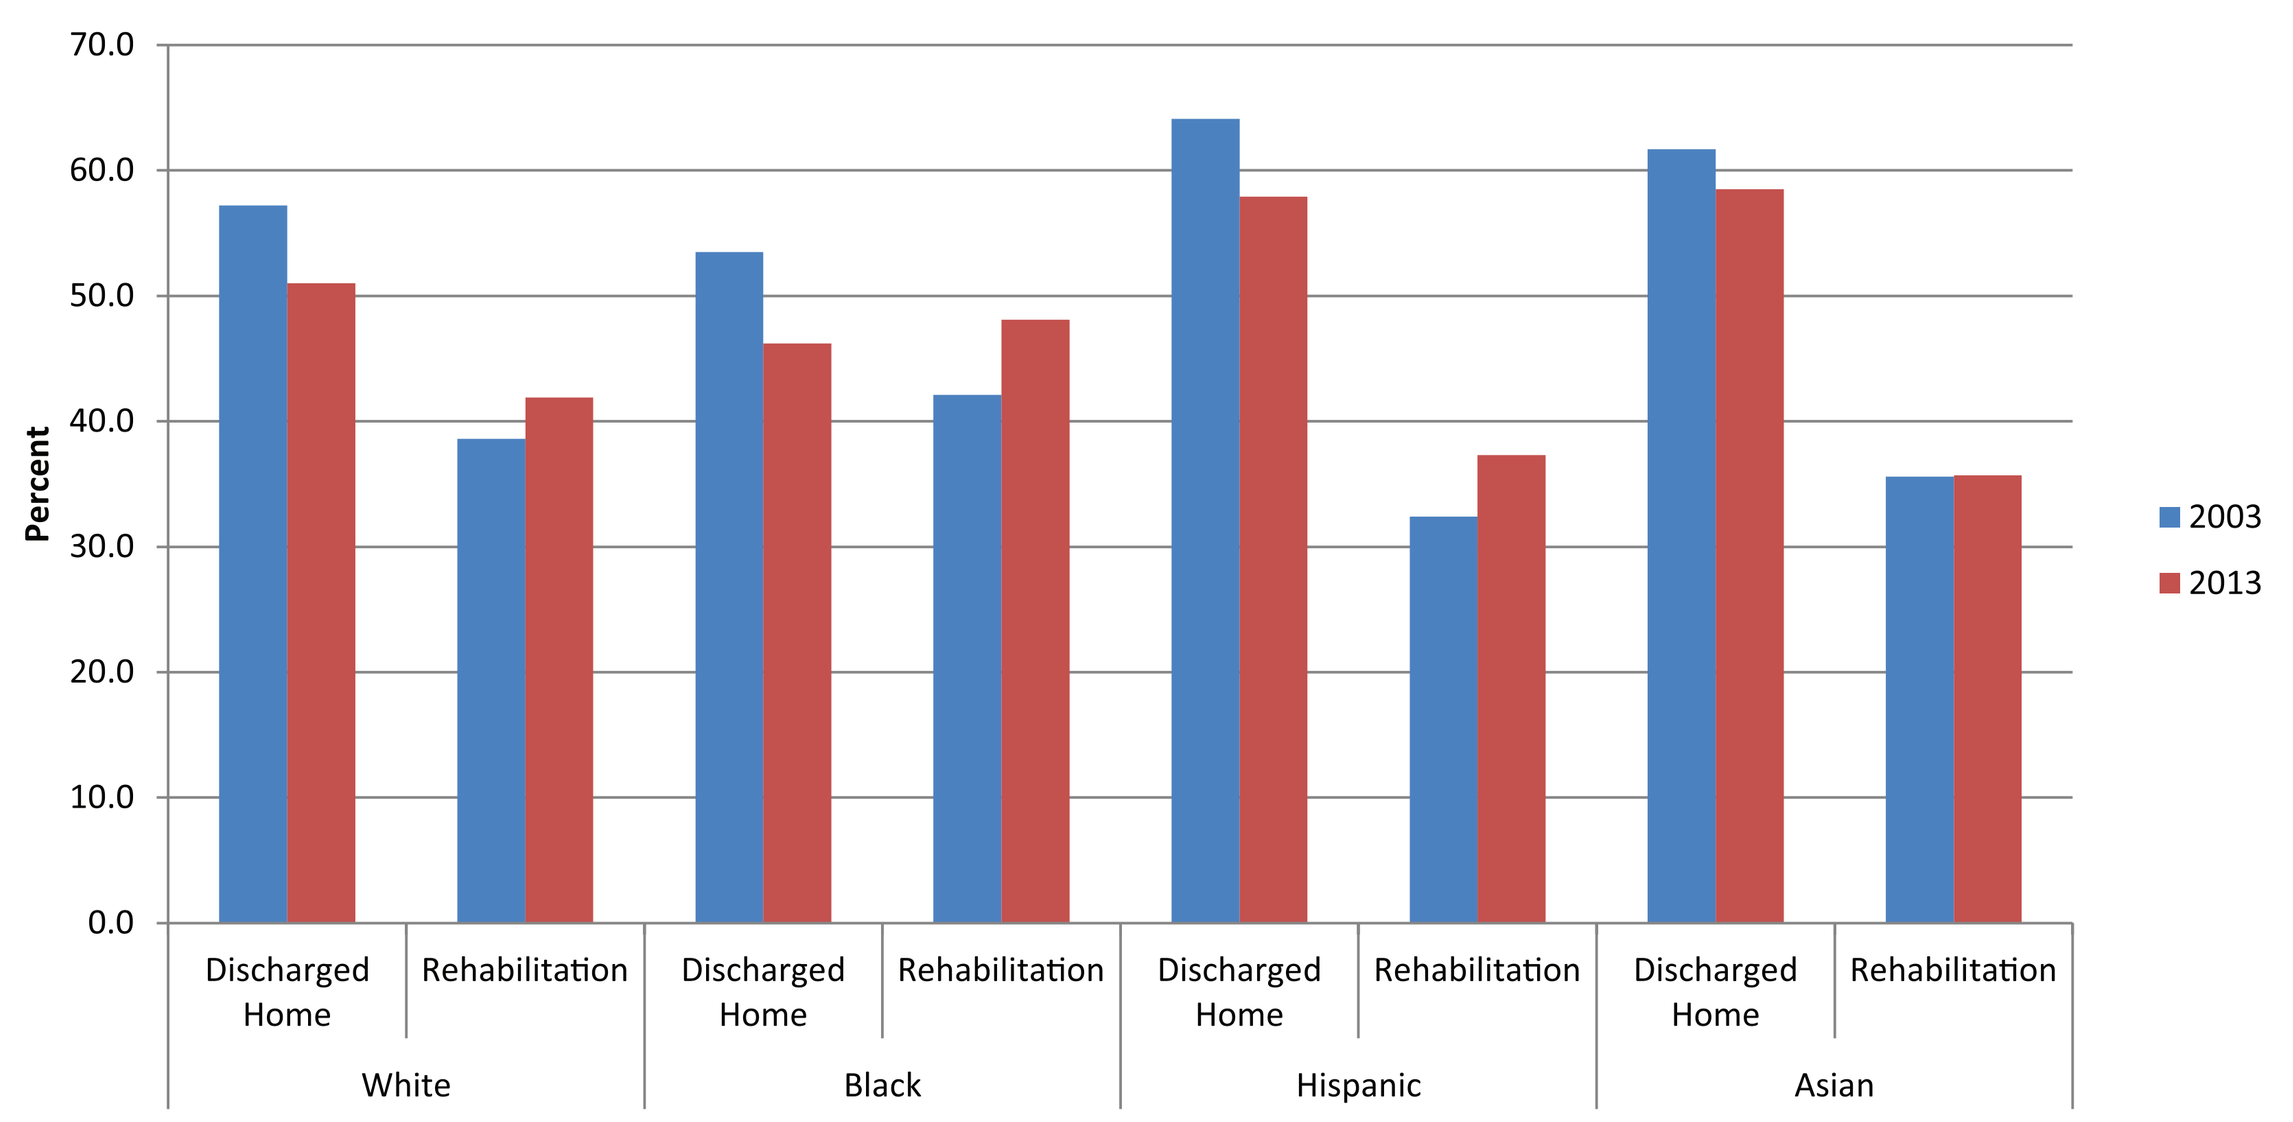

Percent
Discharged Home
Rehabilitation
White
Black
Hispanic
Asian
2003
2013

Trends in Discharge Destination, Length of Stay and Cost of Hospitalization for Ischemic Stroke Patients

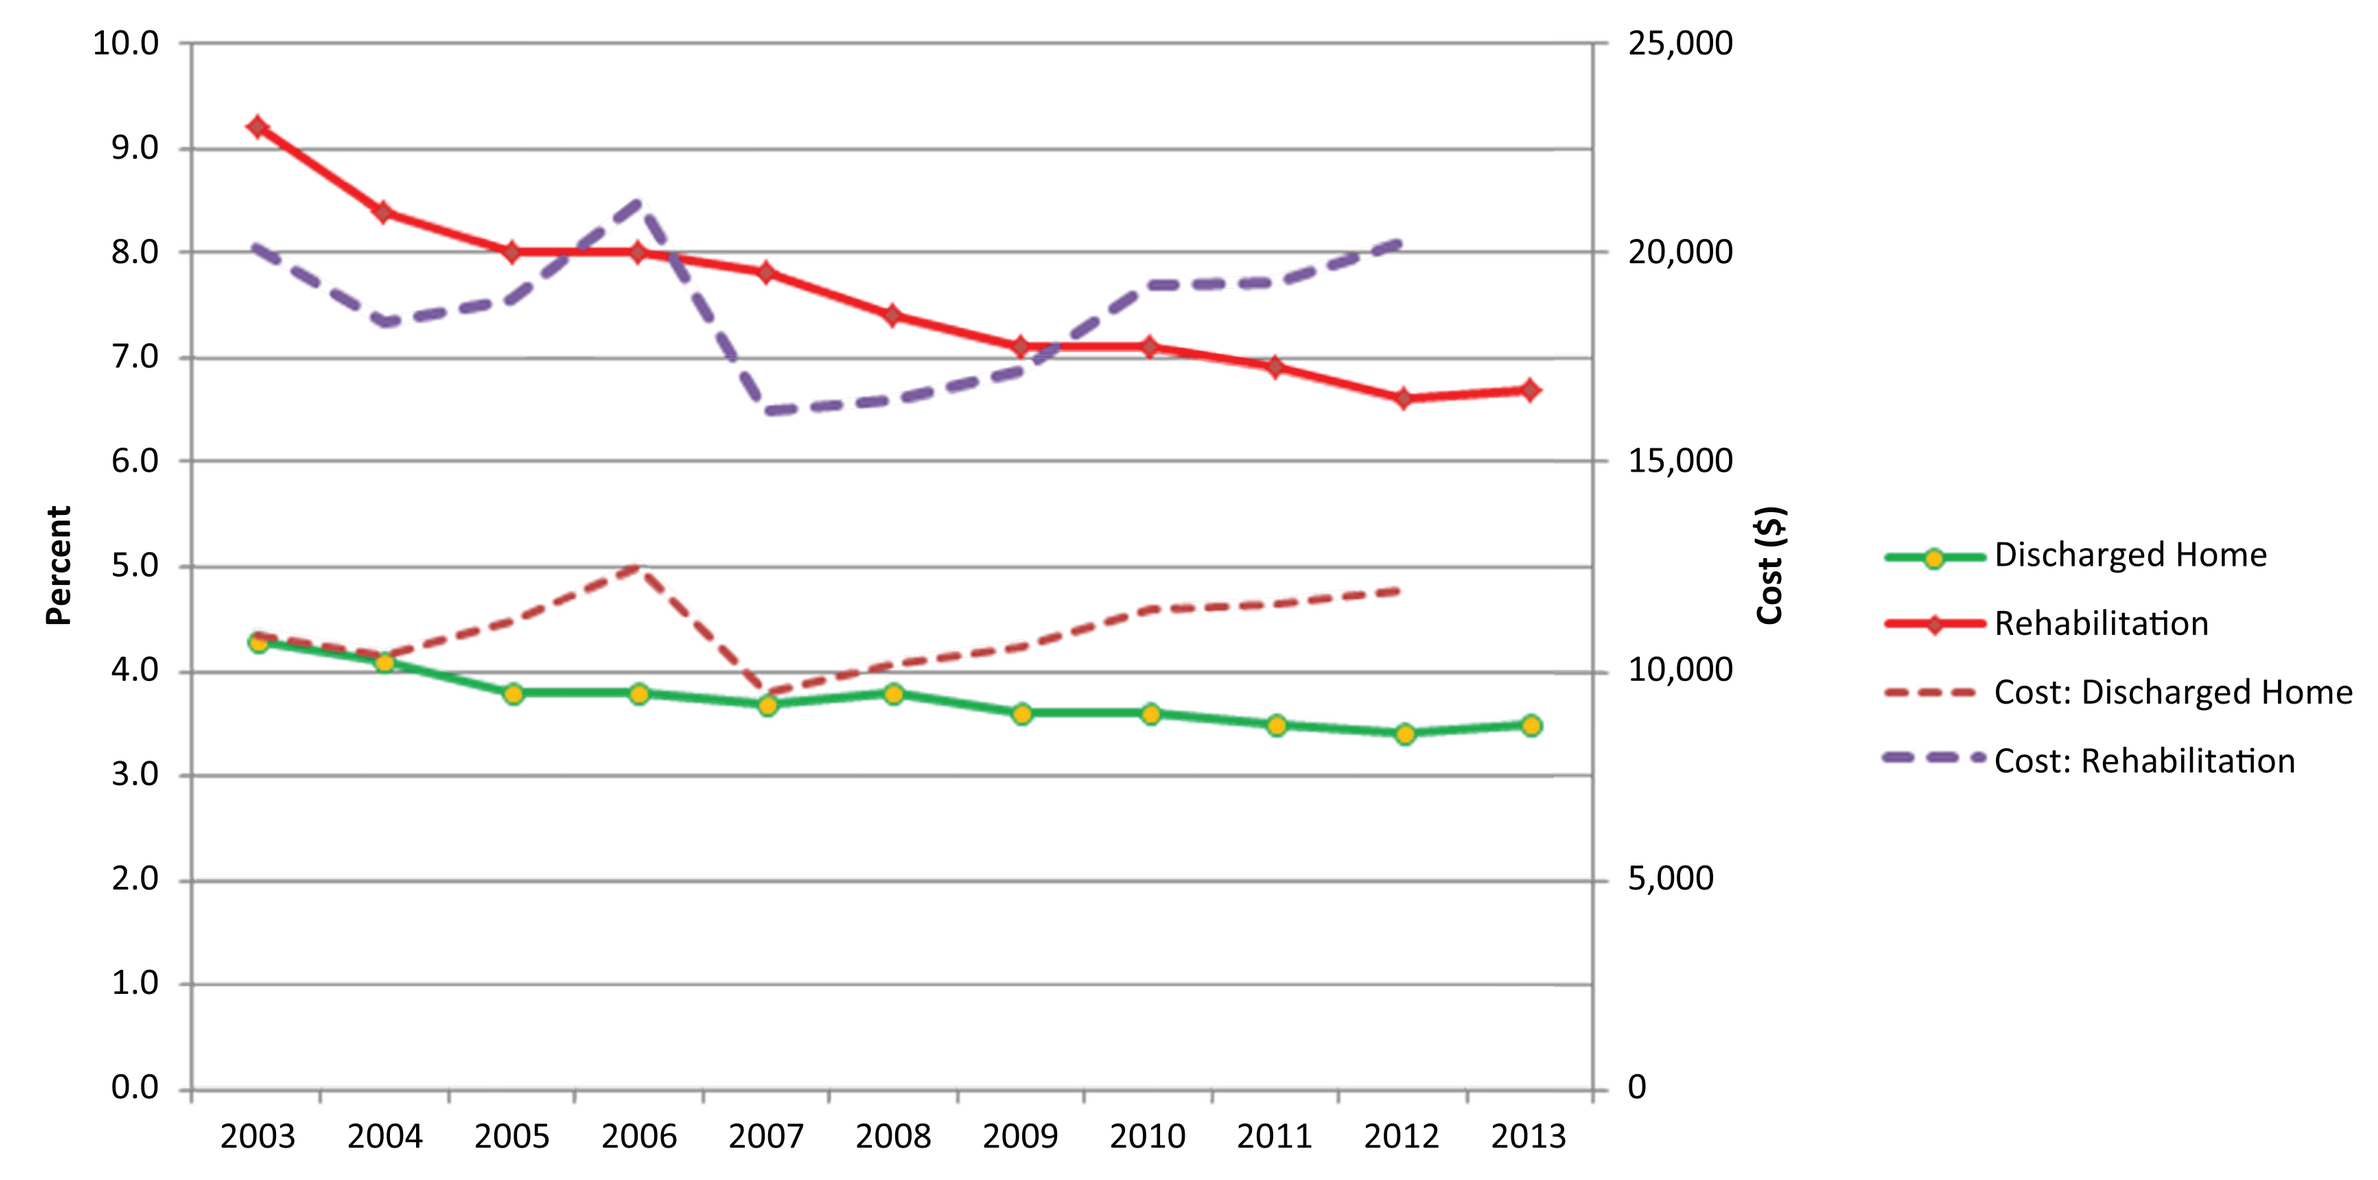

Percent
Cost ($)
2003 2004 2005 2006 2007 2008 2009 2010 2011 2012 2013
Discharged Home
Rehabilitation
Cost: Discharged Home
Cost: Rehabilitation

HCUP Cost Estimator (i.e., cost/charge ratio) not available for 2013

Mean Length of Hospital Stay (LOS) and Average Cost of Hospitalization (Cost) by Type of Stroke and Discharge Destination: 2004, 2008 and 2012

		2004		2008		2012	
Stroke Type	Discharge Destination	LOS	Cost	LOS	Cost	LOS	Cost
Ischemic	Discharged Home	4.1	10,388	3.8	10,176	3.4	11,959
	Rehabilitation	8.4	18,323	7.4	16,482	6.6	20,226
Subarachnoid	Discharged Home	12.0	36,379	11.1	36,828	11.3	50,683
	Rehabilitation	23.2	69,509	17.8	55,443	19.8	92,877
Hemorrhagic	Discharged Home	7.7	18,947	6.6	16,040	5.6	17,862
	Rehabilitation	11.7	28,118	11.5	29,017	9.3	31,492
All Stroke	Discharged Home	4.5	11,396	4.2	11,250	3.9	13,707
	Rehabilitation	9.1	20,473	7.5	22,212	7.4	24,014

Mean Length of Hospital Stay (LOS) in Days, Average Cost of Hospitalization (Cost) by Stroke Type and Race/ethnicity: 2004, 2008, 2012

		2004		2008		2012	
Stroke Type	Race	LOS	Cost	LOS	Cost	LOS	Cost
Ischemic	White/Non-Hisp	5.6	13,005	5.1	12,421	4.6	15,244
	Black/Non-Hisp	7.9	18,600	6.9	17,088	6.1	18,959
	Hispanic	6.8	17,482	6.9	17,226	5.7	16,856
	Asian Non-Hisp	6.4	14,763	6.0	14,341	5.5	16,508
	All	6.0	14,256	6.0	13,679	5.0	16,296
Subarachnoid	White/Non-Hisp.	10.5	32,385	8.9	27,764	10.8	52,911
	Black/Non-Hisp.	13.8	45,804	10.1	37,708	13.0	69,405
	Hispanic	12.6	38,832	12.8	42,396	14.0	60,780
	Asian Non-Hisp.	12.3	42,254	14.8	46,240	13.6	68,907
	All	12.0	36,656	10.0	33,943	12.0	59,957
Hemorrhagic	White/Non-Hisp.	7.4	18,800	7.1	18,175	6.4	22,026
	Black/Non-Hisp.	10.4	28,529	11.0	31,890	8.6	33,001
	Hispanic	13.7	33,343	7.9	21,371	7.5	23,817
	Asian Non-Hisp.	9.6	31,794	10.1	24,823	8.8	33,700
	All	9.0	22,634	8.0	21,645	7.0	25,440
All Stroke	**All Races**	**7.0**	**15,968**	**6.0**	**15,412**	**6.0**	**19,325**

Results

- The 65 or older consistently account for more than 70.0% of all ischemic stroke hospitalization in the State.
- Among both 65+ and 45-64 years old ischemic stroke patients, black or African Americans have the highest percentage discharged to rehabilitation compared to other race/ethnic groups.
- Over 65.0% of all acute stoke patients had Medicare coverage with private insurance accounting for about 23.0%.
- The indigent, self pay and private insured ischemic stroke patients tend to be discharged home at a higher proportion than others.
- Average length of stay for ischemic stroke patients discharged home declined from 4.3 days in 2003 to 3.5 days in 2013. For those discharged to rehabilitation facilities, length of stay declined from 9.2 in 2003 to 6.7 in 2013 while cost of hospitalization per patient increased persistently between 2007 and 2012.
- Even after primary and comprehensive stroke designation, the distributions of ischemic patient discharges by designation status has not changed much, suggesting that there is no strong evidence to support that designation of hospitals has made a measurable influence on the patterns of discharge destination.
- However, a test of relationship between discharge destination and Modified Rankin scale using 2013 NJASR (after excluding those who died) shows that the relationship was statistically significant ($\chi 2(15) < 0.0001$).

Policy Implications and Recommendations

- There is an important difference by race/ethnicity in ischemic stroke hospitalization which has persisted over the study period.
- Stroke continues to be a major health problem whose risk factors can be modified through effective and targeted public education.
- Further research that focuses on stroke risk factors associated with population groups need to be conducted to support health policy making.
- With most hospitals in New Jersey designated as stroke centers, a careful analysis of the implications of designating hospitals as stroke centers on stroke treatment outcomes is important.

References

- Hospitalizations for Acute Stroke in New Jersey: Trends in Volume, Mortality and Patient Demographics **http://www.nj.gov/health/healthcarequality/stroke/reports.shtml**
- Health Care Quality Assessment; *Inpatient Quality Indicators Reports*: **http://www.nj.gov/health/healthcarequality/qi.shtml**
- Stroke Services: **http://www.nj.gov/health/healthcarequality/stroke/index.shtml**
- The New Jersey Acute Stroke Registry: **http://www.nj.gov/health/healthcarequality/stroke/initiatives.shtml**
- Summary Stroke Statistics: **http://www.nj.gov/health/healthcarequality/stroke/statistics.shtml**
- The New Jersey Hospital Data Collection System: **http://www.nj.gov/health/healthcarequality/hospitaldata.shtml**